LC/MS Charge Variant Characterization of an ECD-Fc Fusion Protein using OFFGEL and SAX Fractionation Workflows

Tim Keefe, Jamie Strand, Alan Akiyama, Chi-Ting Huang

Acceleron Pharma, Cambridge, MA

Abstract

Receptor extracellular domain fusion proteins represent a growing class of biotherapeutics. Since these fusion proteins can be highly glycosylated they have broad distributions of charge variants. In todays biotechnology industry there is an increasing need to structurally characterize the charge heterogeneity of these variants by methods other than electrophoresis. In this study we have developed rigorous analytical workflows to structurally characterize an ECD-Fc fusion protein using both pI based separation (OFFGEL fractionation) and IEX chromatography to fractionate the protein heterogeneity upstream. We have found that the glycosylation in the ECD region is the major contributor to the overall charge heterogeneity. These glycosylated species have been fractionated and structurally characterized using OFFGEL fractionation and Strong Anion Exchange (SAX) chromatography workflows followed by LC/MS.

Introduction

Fc fusion proteins, a growing class of biological therapeutic, are challenging to characterize because of their heterogeneity, especially for highly glycosylated ECD-Fc fusion proteins. Charge heterogeneity of highly glycosylated biological therapeutics is being monitored more frequently using techniques such as IEF, cIEF, and IEC. Each of these techniques can result in very broad bands and a distribution of charge and pI values that are difficult to resolve and structurally characterize.

OFFGEL fractionation is an isoelectric focusing technique that has been specifically designed by Agilent Technologies so that proteins or peptides can be recovered in solution upstream of LC/MS characterization. In this study the OFFGEL fractionator is employed as the basis for the pI based workflow where we have fractionated the protein and structurally characterized the charge variants downstream by LC/MS.

Anion Exchange chromatography has been widely used to characterize the charge heterogeneity profile of Mabs for developing therapeutics to ensure product quality and consistency during processing. Resolving charge variants of an ECD-Fc fusion protein can be more challenging mainly because the glycan rich ECD domain contributes a more heterogeneous profile. In this study a robust pH based AEX method has been developed to resolve charge variants as part of a workflow to characterize the fractions downstream by LC/MS.

Methods

OFFGEL fractionation:
An Agilent OFFGEL 3100 fractionator was used along with pH 4-7 Immobiline DryStrips from GE Healthcare to fractionate 3mg of protein per lane in a 24 well frame. Following the fractionation adjacent wells were combined into 12 fractions.

Strong Anion Exchange Chromatography
A non-porous quaternary amine column (Proteomics SAX-NP3; 3um; 7.8mm x 150mm) from SEPAX Technologies was used to fractionate 150 ug protein per injection. An Agilent 1100 HPLC system was used to pump the pH gradient as described in Figure 7.

LC/MS
- OFFGEL and SAX fractions were desalted prior to LC/MS analysis using Amicon Ultra 10K Centrifugal filters.
- Peptide mapping was performed on a Jupiter 4um Proteo 90A, 4.6mm x 250mm. Buffer A was 0.05%TFA and bufer B was 0.05% TFA in acetonitrile. An Agilent 1200 was used to deliver a gradient of 2% to 35% in 100 minutes. A Thermo LTQ-Orbitrap-XL acquired MS data at 60k resolution for full MS scans and CID of the top 3 ions were acquired in the LTQ.
- Subunit LC/MS data of the fusion protein was acquired by first digesting the protein with the IdeS enzyme (FabRICATOR; Genovis, Lund Sweden). The subunits were denatured with GuHCL followed by reducing the disulfide bonds with DTT and alkylated with iodoacetamide . The resulting subunits were separated on an Xbridge C4 BEH300, 2.1mm x 100mm. A Thermo LTQ-Orbitrap-XL acquired full MS scans at 7500 resolution.

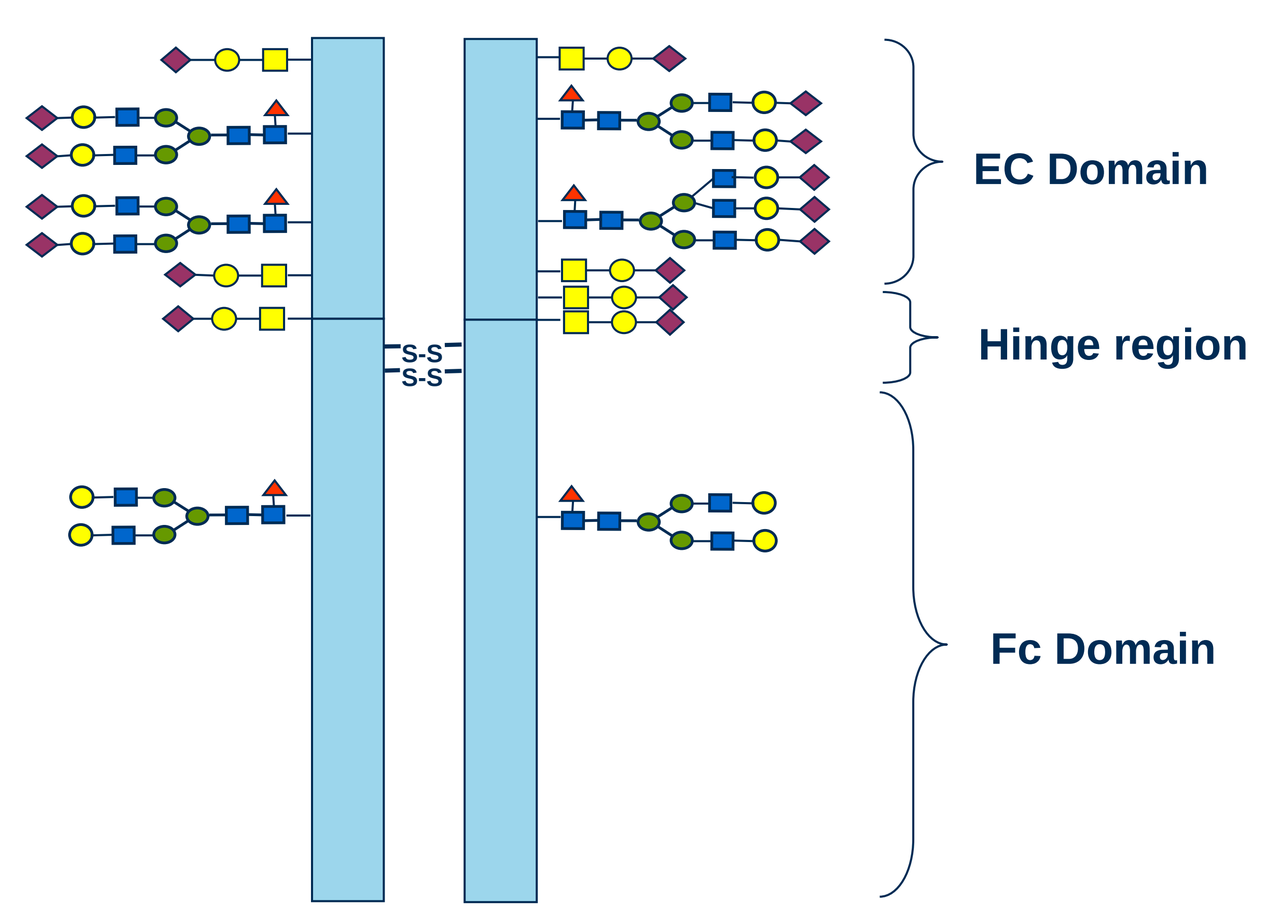

S-S
S-S
EC Domain
Hinge region
Fc Domain

Figure 1. General structure of an ECD-fusion protein

A cartoon of a therapeutic protein construct is shown which fuses an active extracellular domain to the Fc domain of a human IgG. A construct that we are currently developing has several glycosylation sites. There are two N-link glycosylation sites on each monomer of the ECD and one N-link site per monomer on the Fc domain. There are also up to 4 O-link glycosylation sites on each monomer in the ECD region.

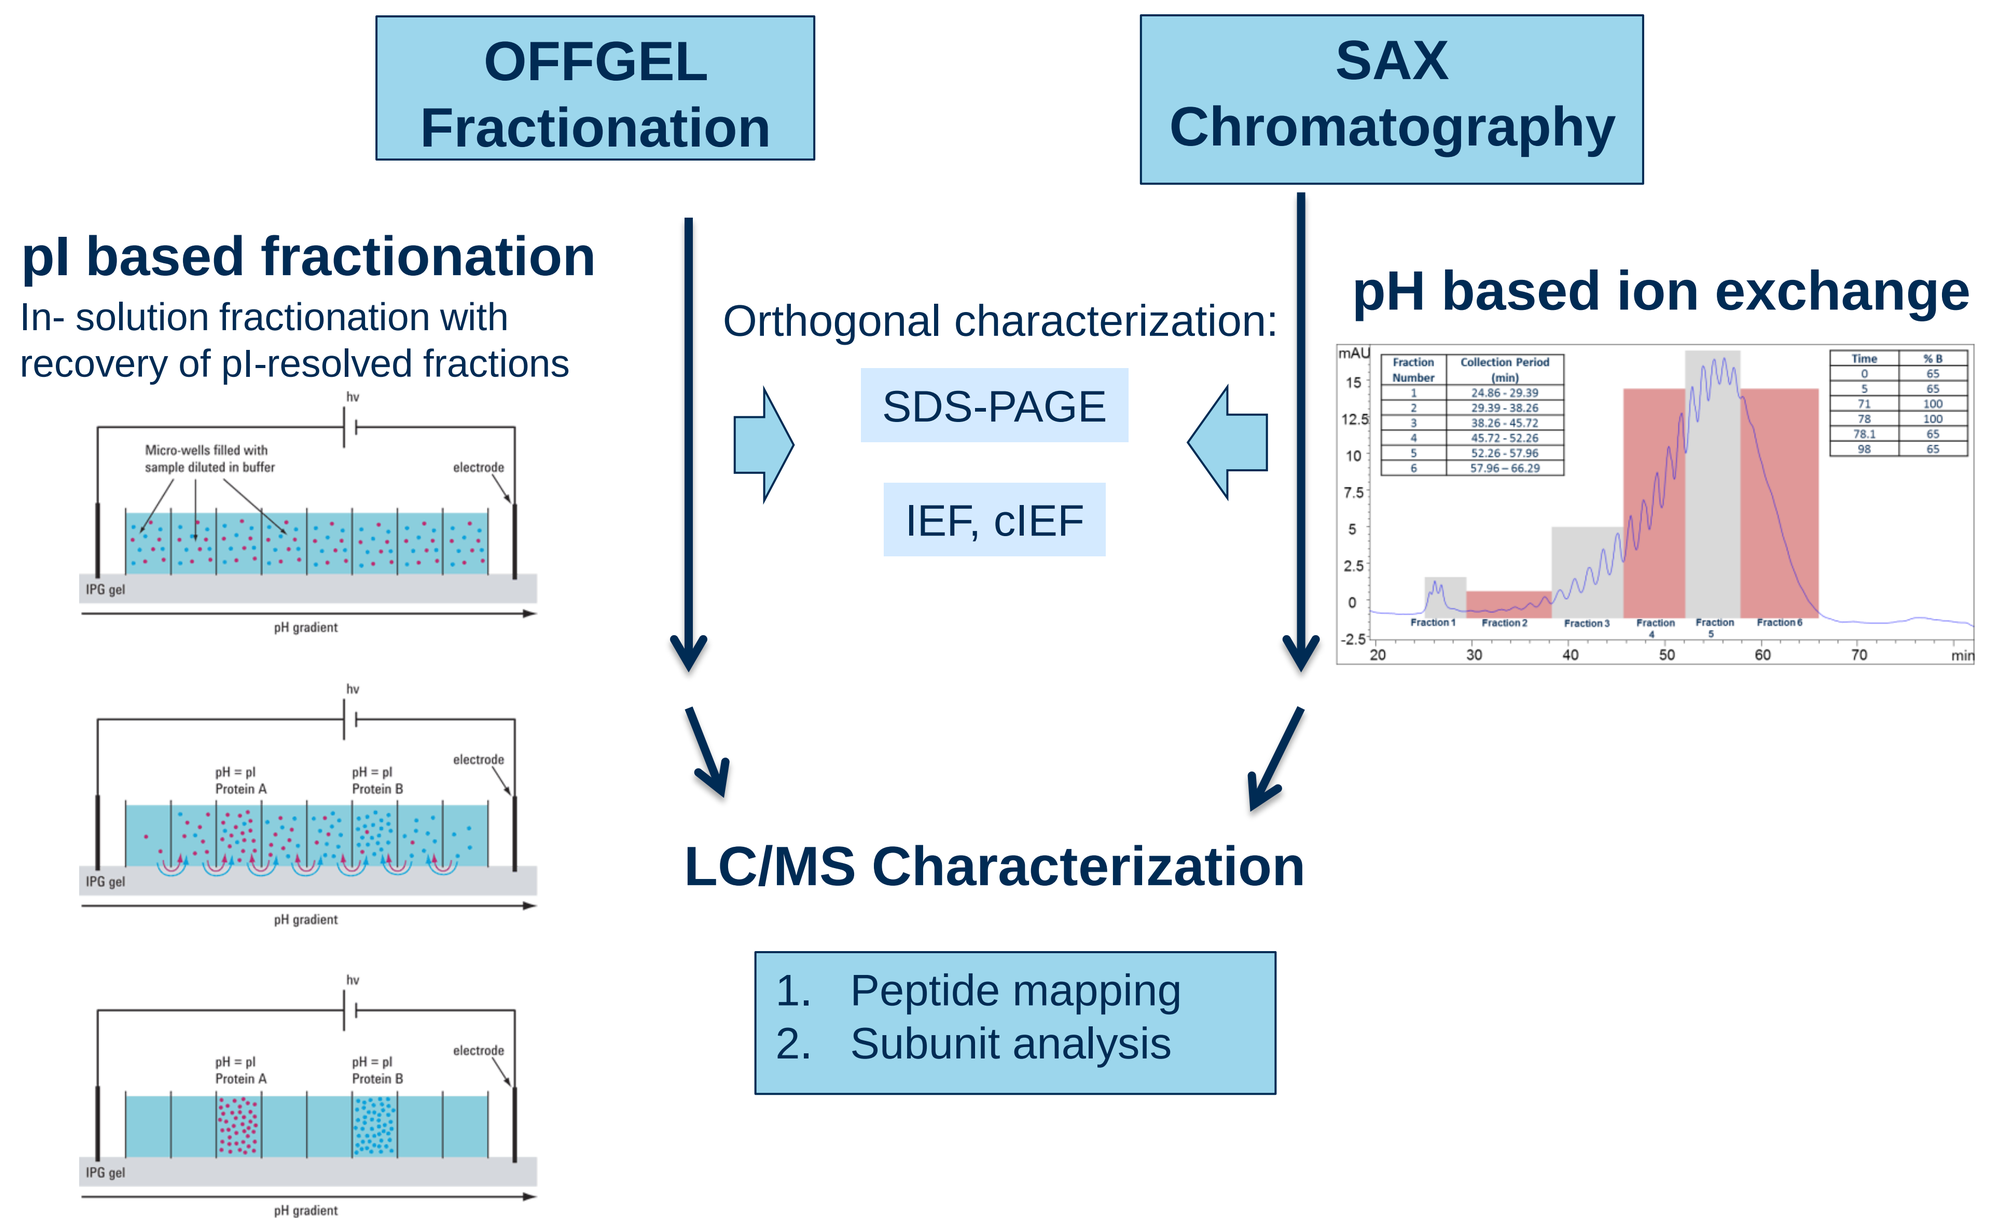

OFFGEL Fractionation
SAX Chromatography
pI based fractionation
In- solution fractionation with recovery of pI-resolved fractions
Orthogonal characterization:
SDS-PAGE
IEF, cIEF
pH based ion exchange
LC/MS Characterization
1. Peptide mapping
2. Subunit analysis

Figure 2. Characterization Workflow

Characterization of the protein charge heterogeneity in this study is based on two fractionation workflows with LC/MS endpoints. One workflow uses the OFFGEL fractionation which is a pI based separation of the protein into wells which are easily recovered in solution after the charged variants are fully focused. The second is strong anion exchange (SAX) chromatography with offline fraction collection.

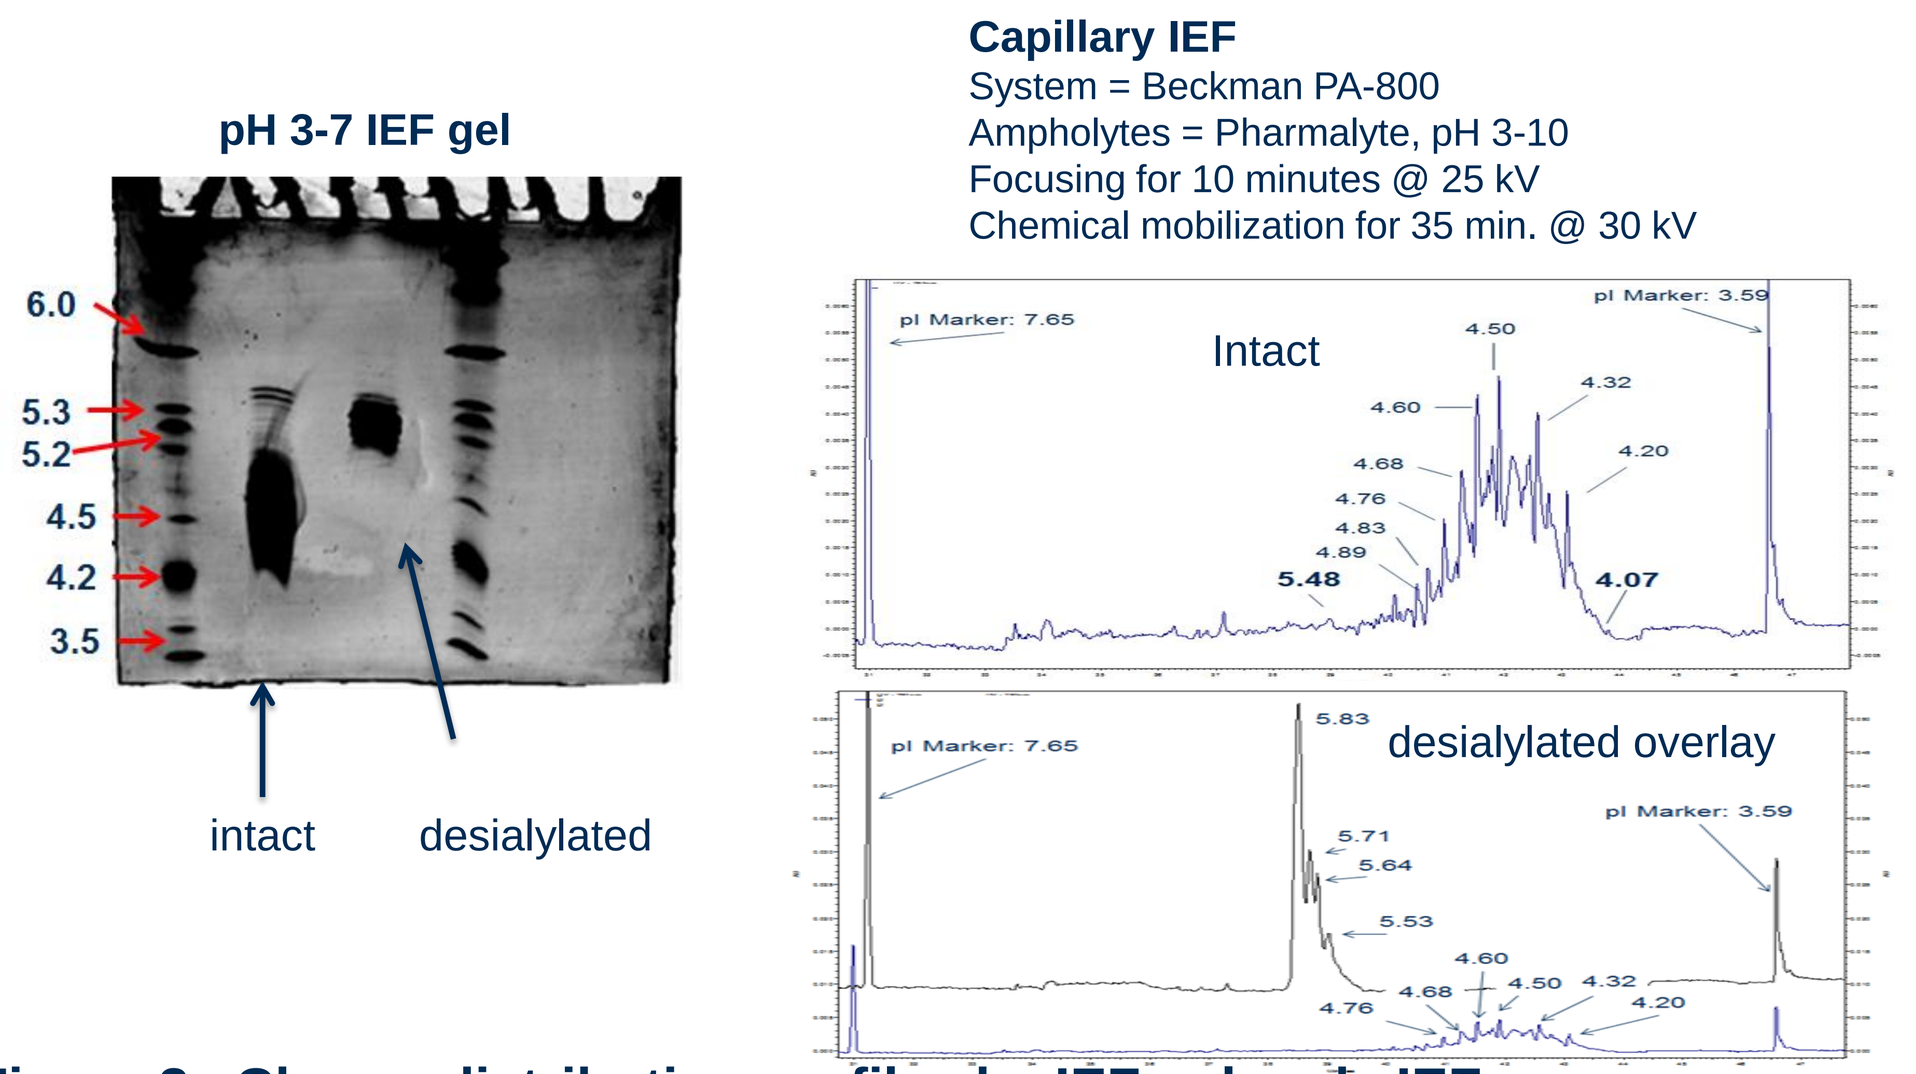

pH 3-7 IEF gel
6.0
5.3
5.2
4.5
4.2
3.5
intact
desialylated
Capillary IEF
System = Beckman PA-800
Ampholytes = Pharmalyte, pH 3-10
Focusing for 10 minutes @ 25 kV
Chemical mobilization for 35 min. @ 30 kV
Intact
desialylated overlay
pI Marker: 7.65
pI Marker: 3.59

Figure 3. Charge distribution profiles by IEF gel and cIEF

Both techniques separate charge variants according to their pI values and are critical for monitoring batch to batch variability. The capillary technique has advantages in the analysis of highly charged fusion proteins such as resolution and its quantitative aspects. However, both techniques are difficult to adapt into further characterization workflows for structural identification.

OFFGEL Fraction Characterization

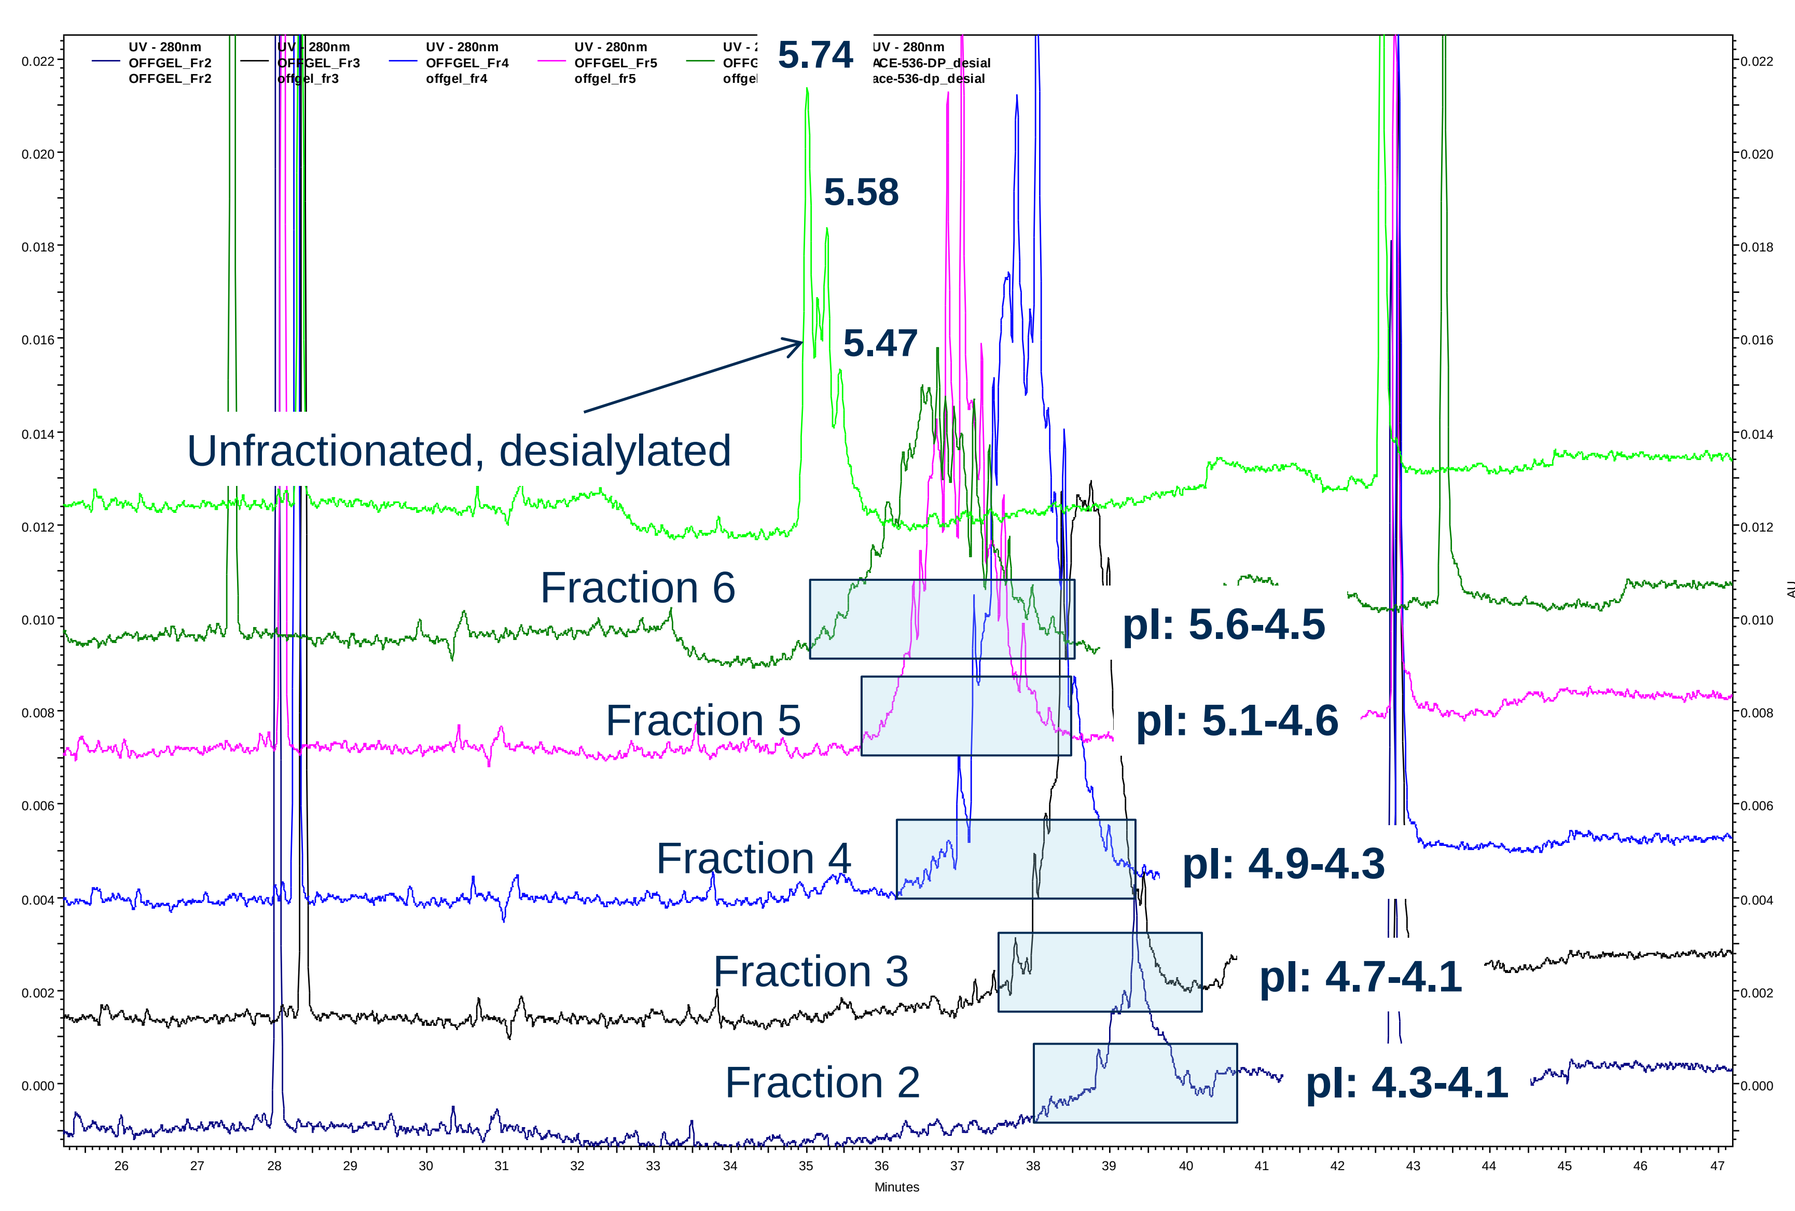

5.74
5.58
5.47
Unfractionated, desialylated
Fraction 6
Fraction 5
Fraction 4
Fraction 3
Fraction 2
pI: 5.6-4.5
pI: 5.1-4.6
pI: 4.9-4.3
pI: 4.7-4.1
pI: 4.3-4.1
Minutes

Figure 4. cIEF profiles of OFFGEL fractions

There are small pI shifts (<0.5 pI units) in each of the OFFGEL fractions showing good resolution of the charge variants. The data shows that at the earlier fractions closest to the anode the protein has lower pI values, as we would expect. These fractions are probably more highly sialylated. The fractions furthest from the anode (fractions 5 and 6) more closely resemble the desialylated control.

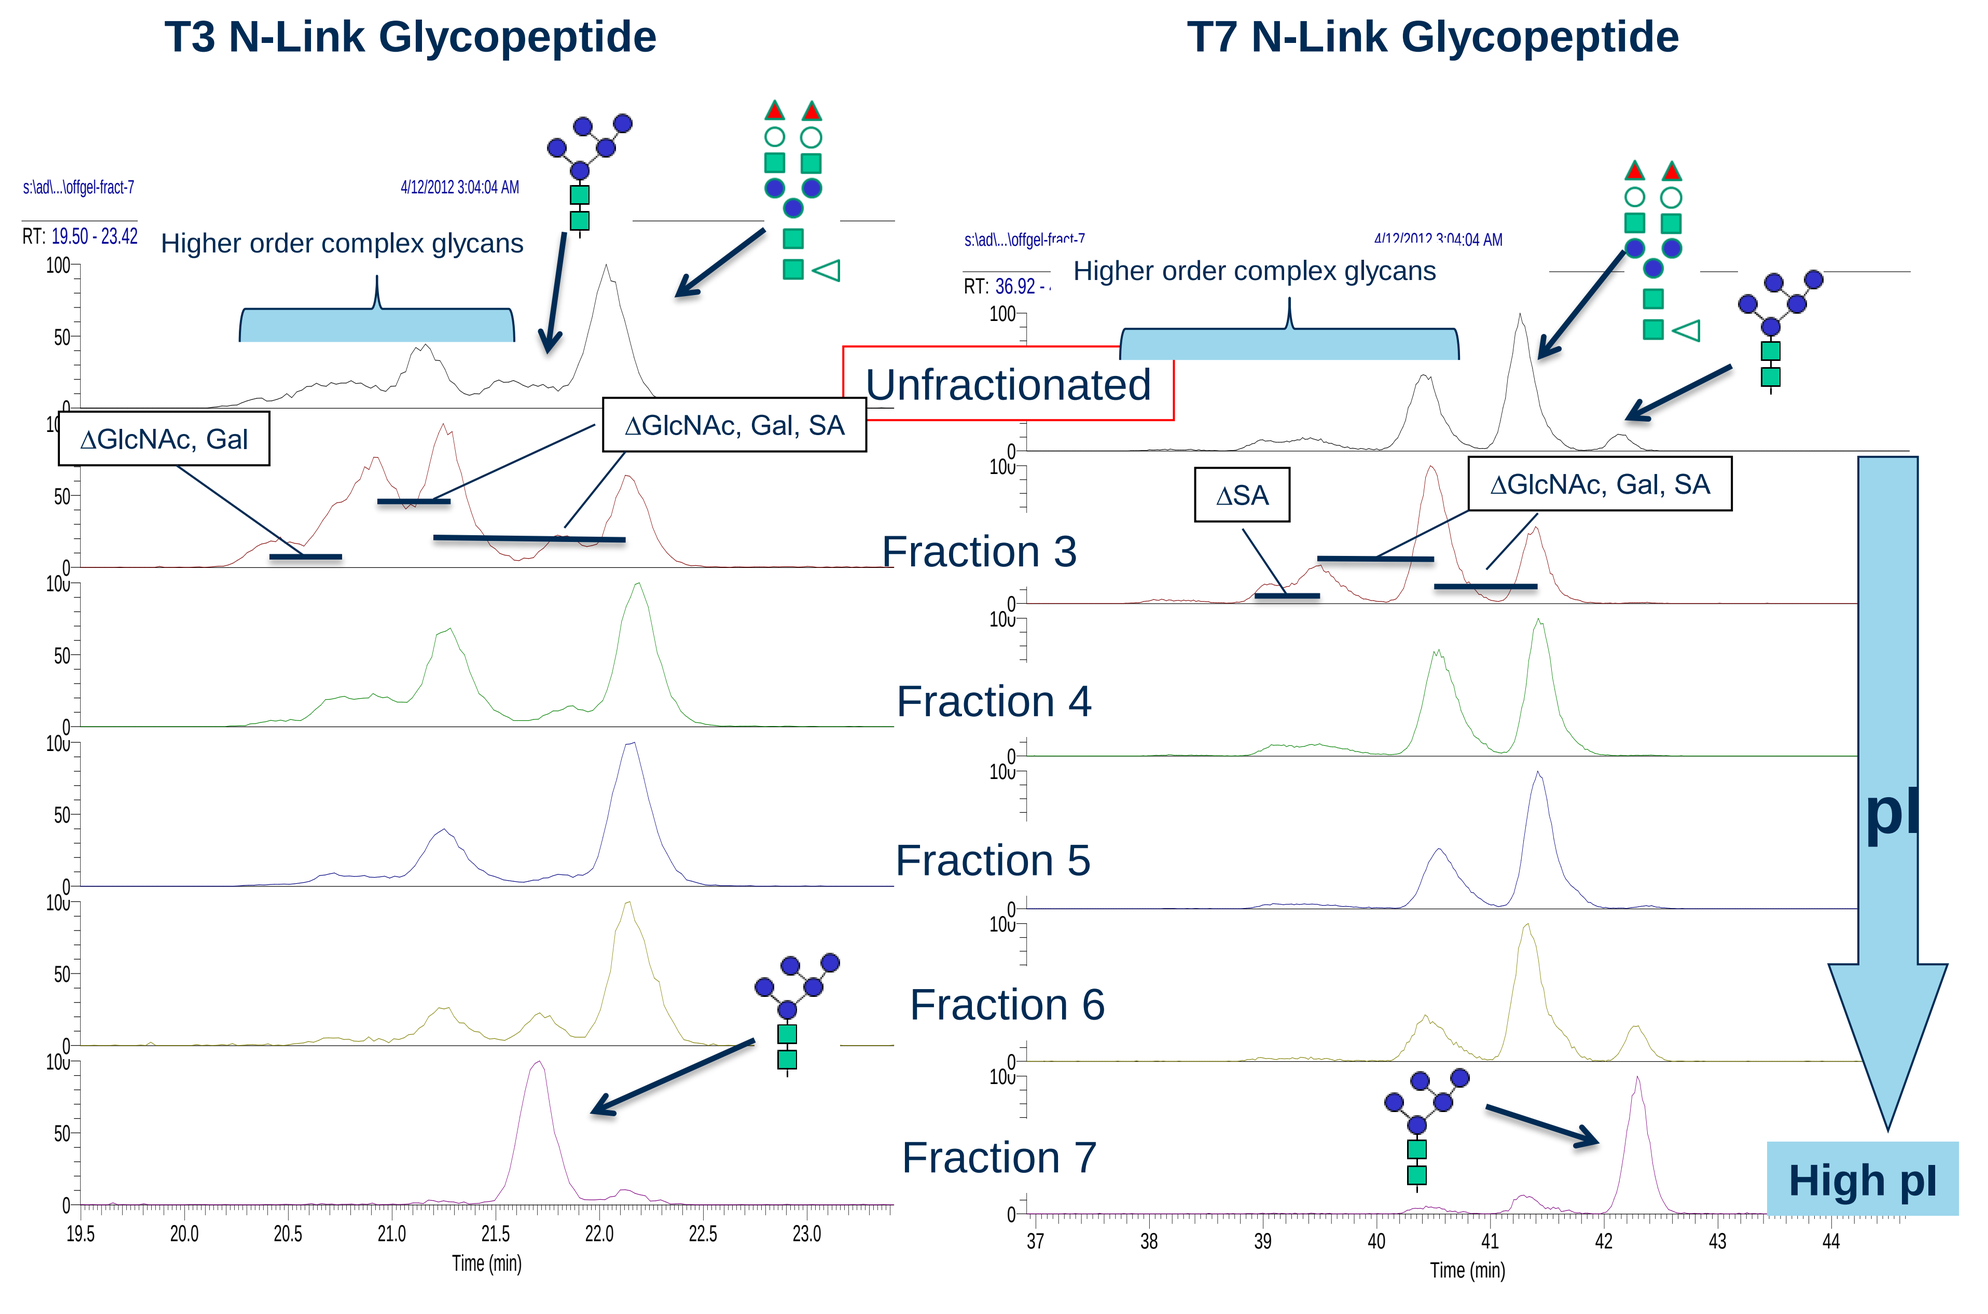

T3 N-Link Glycopeptide
T7 N-Link Glycopeptide
Higher order complex glycans
ΔGlcNAc, Gal
ΔGlcNAc, Gal, SA
ΔSA
Unfractionated
Fraction 3
Fraction 4
Fraction 5
Fraction 6
Fraction 7
pI
High pI
Time (min)

Figure 5. Peptide map analysis of OFFGEL fractions

The two N-link sites of the unfractionated material are similar in glycan structure. The majority of the peptides are occupied with biantennary, fully sialylated glycans. As the pI of the fractions increase, the complexity of the glycans decrease. The majority of fraction 7 is occupied with high mannose glycans on both sites.

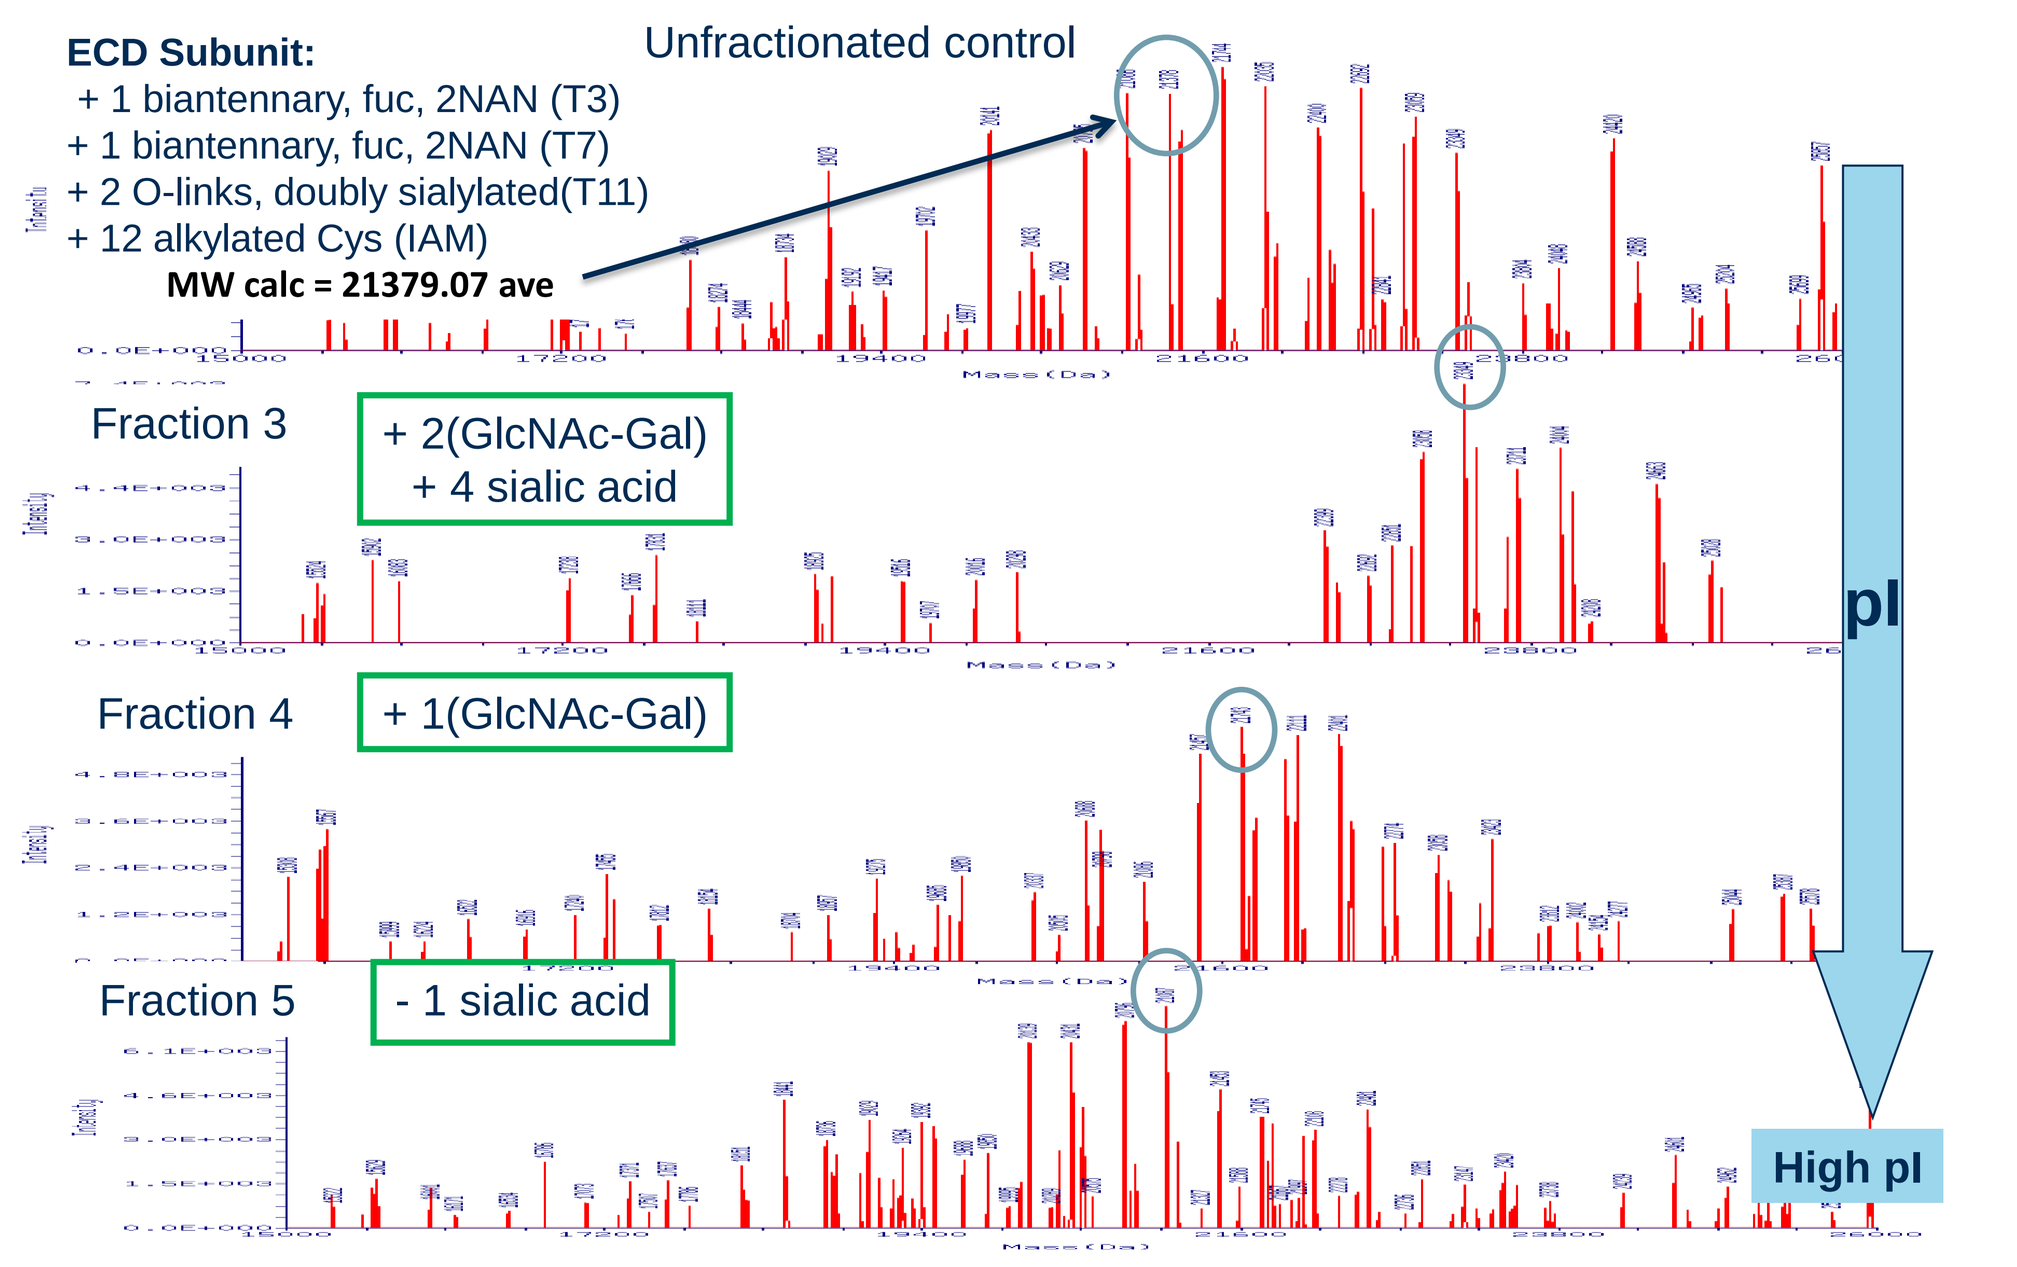

ECD Subunit:
+ 1 biantennary, fuc, 2NAN (T3)
+ 1 biantennary, fuc, 2NAN (T7)
+ 2 O-links, doubly sialylated(T11)
+ 12 alkylated Cys (IAM)
MW calc = 21379.07 ave
Unfractionated control
Fraction 3
+ 2(GlcNAc-Gal)
+ 4 sialic acid
Fraction 4
+ 1(GlcNAc-Gal)
Fraction 5
- 1 sialic acid
pI
High pI

Figure 6. Middle down LC/MS analysis of ECD subunit from OFFGEL fractions

The ECD subunit masses reveals that in the lower pI fractions the glycan structures are more complex and have higher order branching structures with more sialic acid.

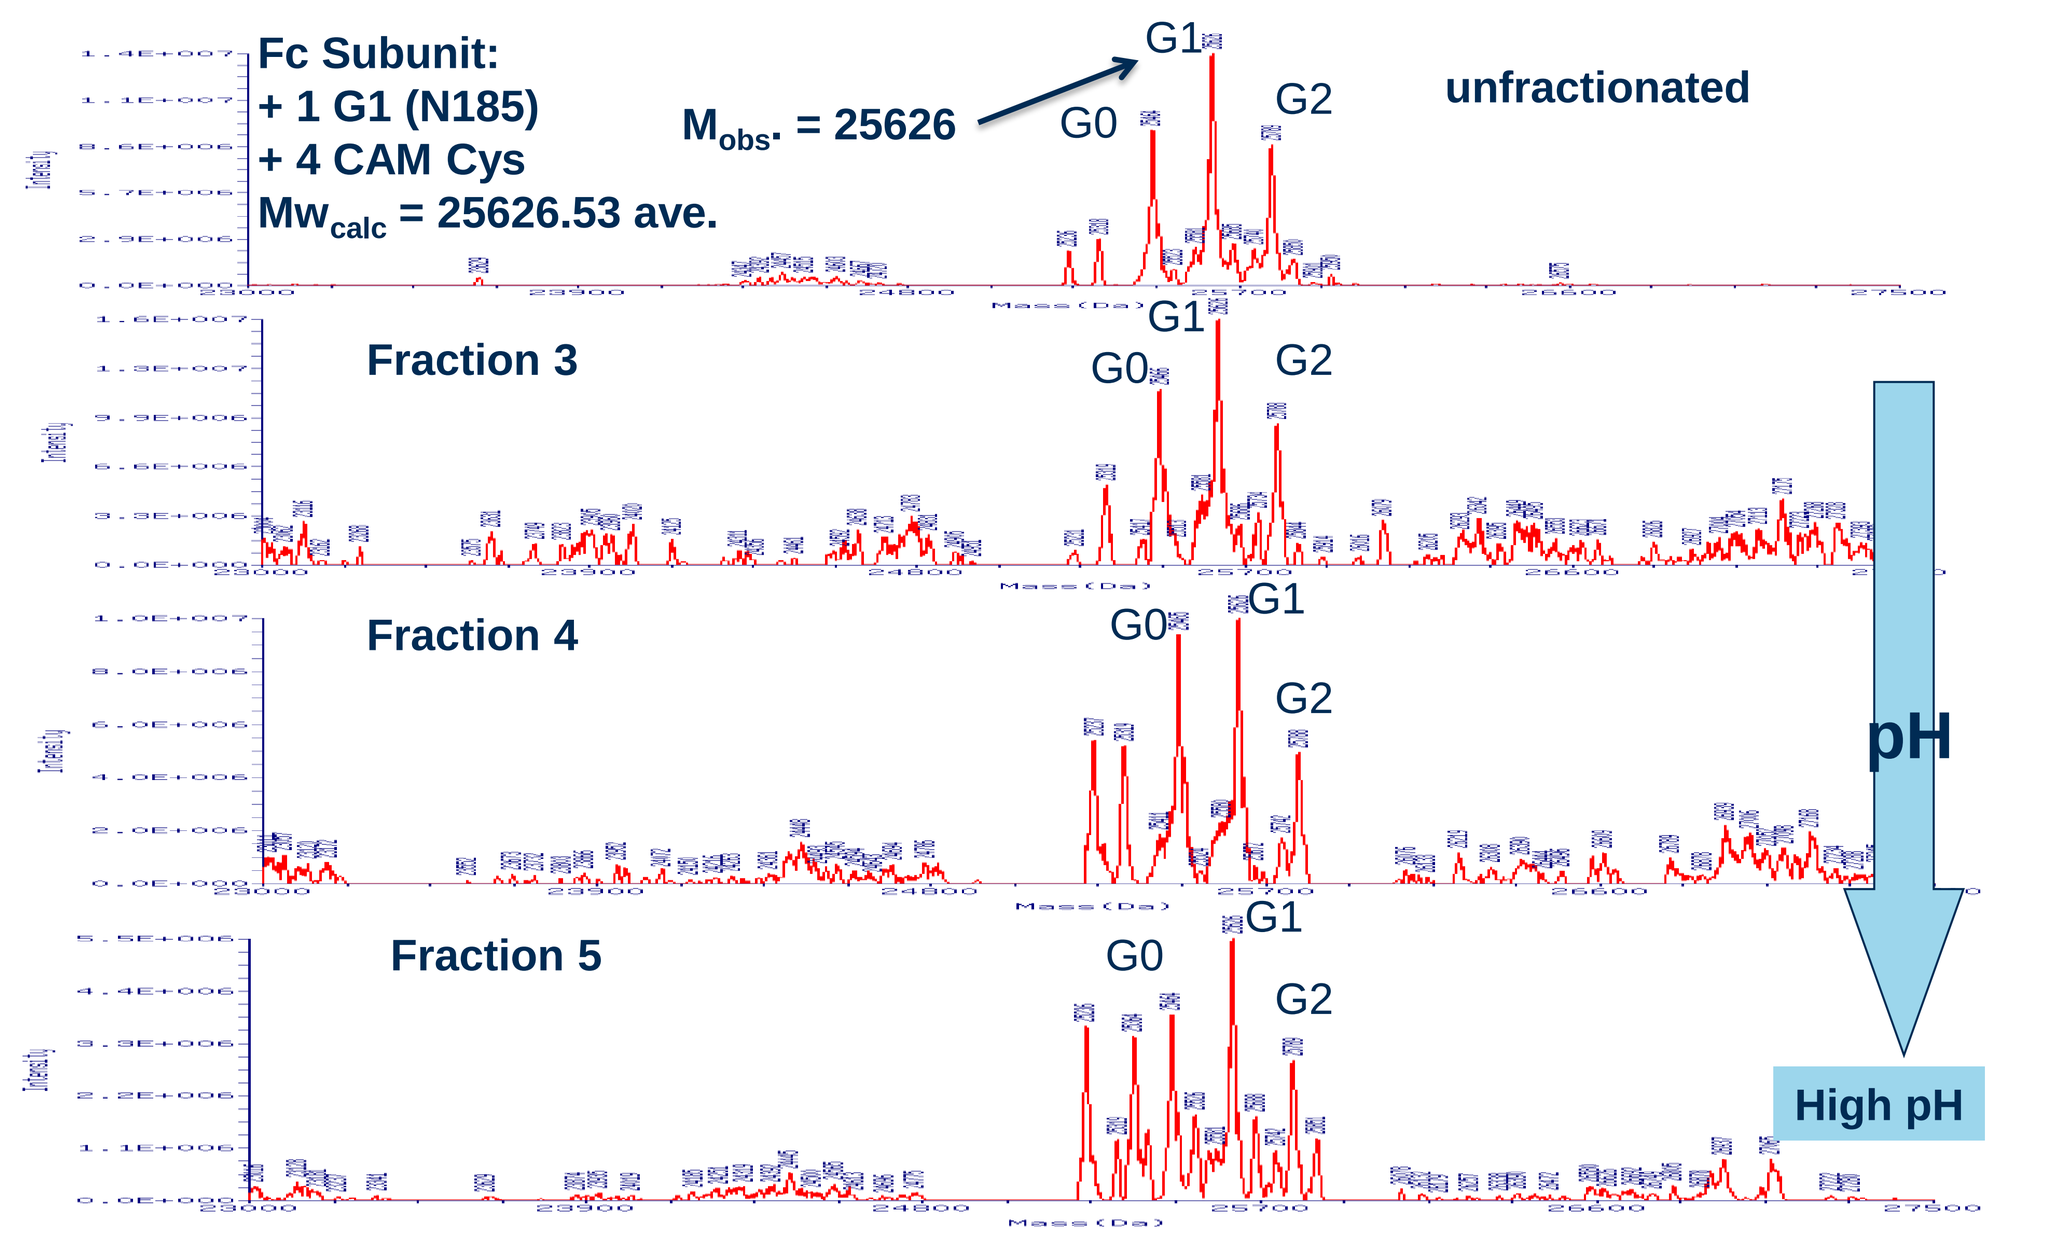

Fc Subunit:
+ 1 G1 (N185)
+ 4 CAM Cys
Mw_{calc} = 25626.53 ave.
M_{obs}. = 25626
G0
G1
G2
unfractionated
Fraction 3
Fraction 4
Fraction 5
pH
High pH

Figure 7. Middle down LC/MS analysis of the Fc domain from the OFFGEL fractions

The Fc domain contains one glycosylation site that is relatively simple compared to the glycans of the ECD domain. We can see that the OFFGEL fractionation has little effect on the glycan distribution and the overall Fc structure.

SAX Fraction Characterization

Column: Proteomix SAX NP3 7.8x150mm (SN: 34120), Mobile phase: A: 20mM Piperazine + 20 mM triethanolamine+20mM bis-tris propane+20mM N-methylpiperazine pH 9.7 (titrate with HCl), B: A at pH 3.7, Flow rate: 1.0 ml/min, Detection: UV 280 nm, Sample: ACE-536 (5 mg/mL), Injection: 10 μL, Column temperature: 50°C

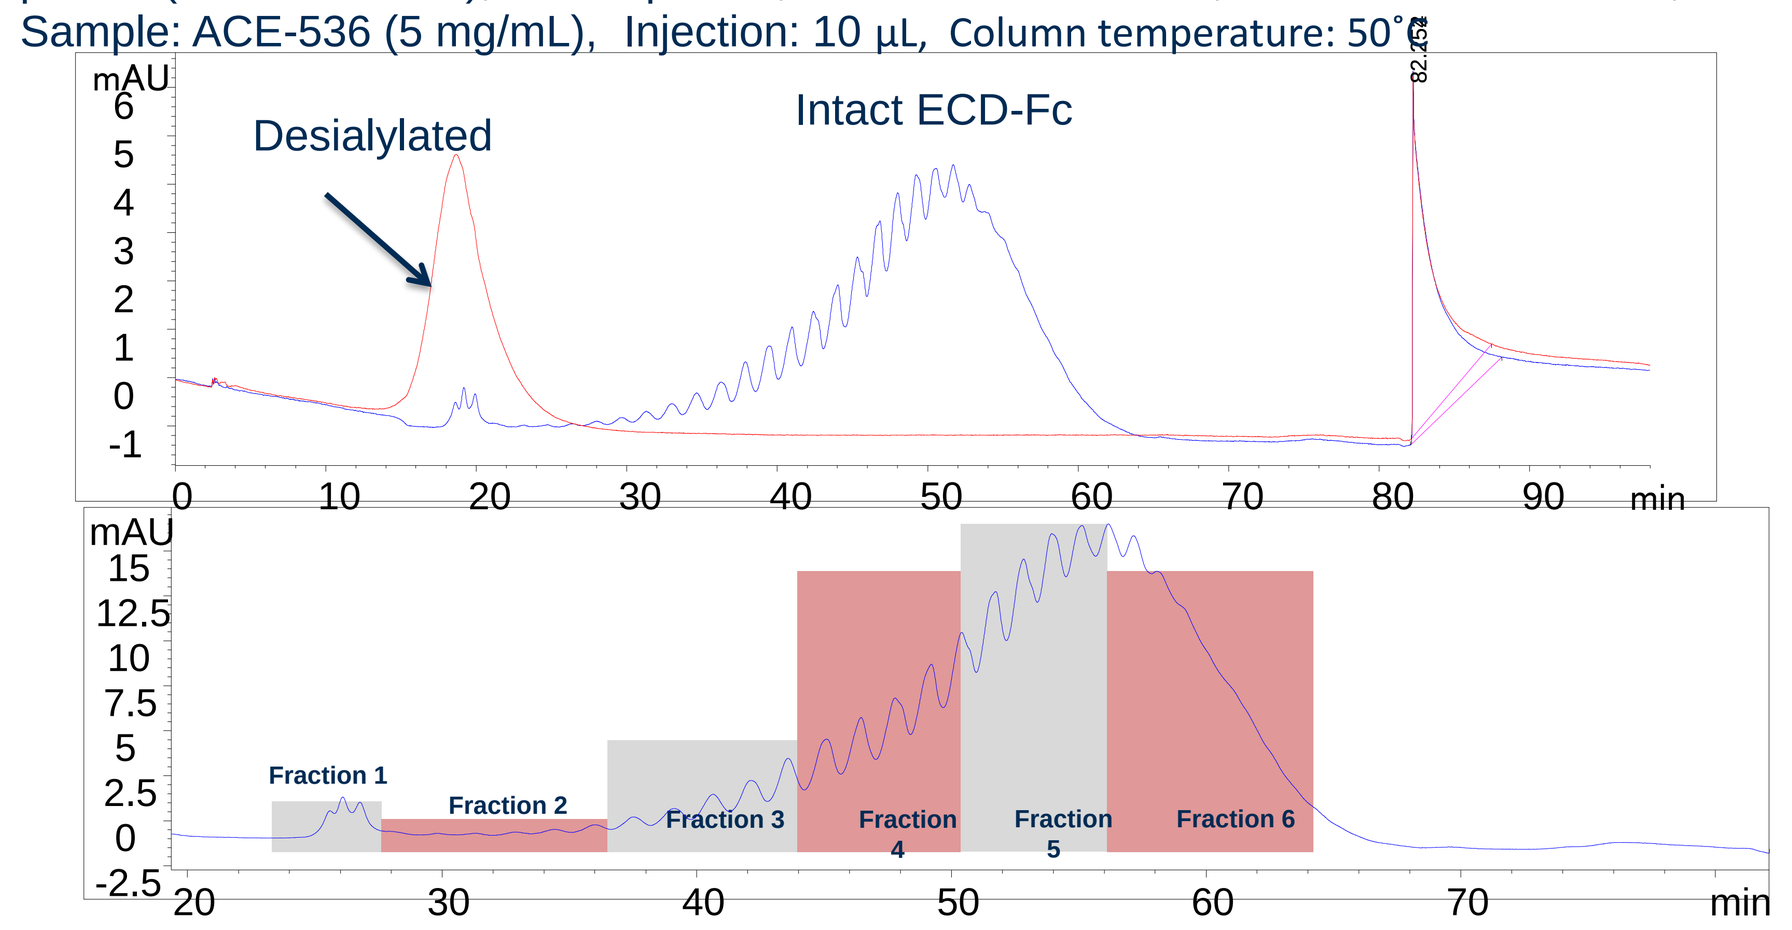

Desialylated
Intact ECD-Fc
mAU
min
Fraction 1
Fraction 2
Fraction 3
Fraction 4
Fraction 5
Fraction 6

Figure 8. SAX Chromatography of the ECD fusion protein

Strong Anion Exchange (SAX) is used here to separate charge variants using a pH based mobile phase on a quaternary amine stationary phase. We can see in the top trace that the early eluting species at about 20 minutes is consistent with the desialylated protein.

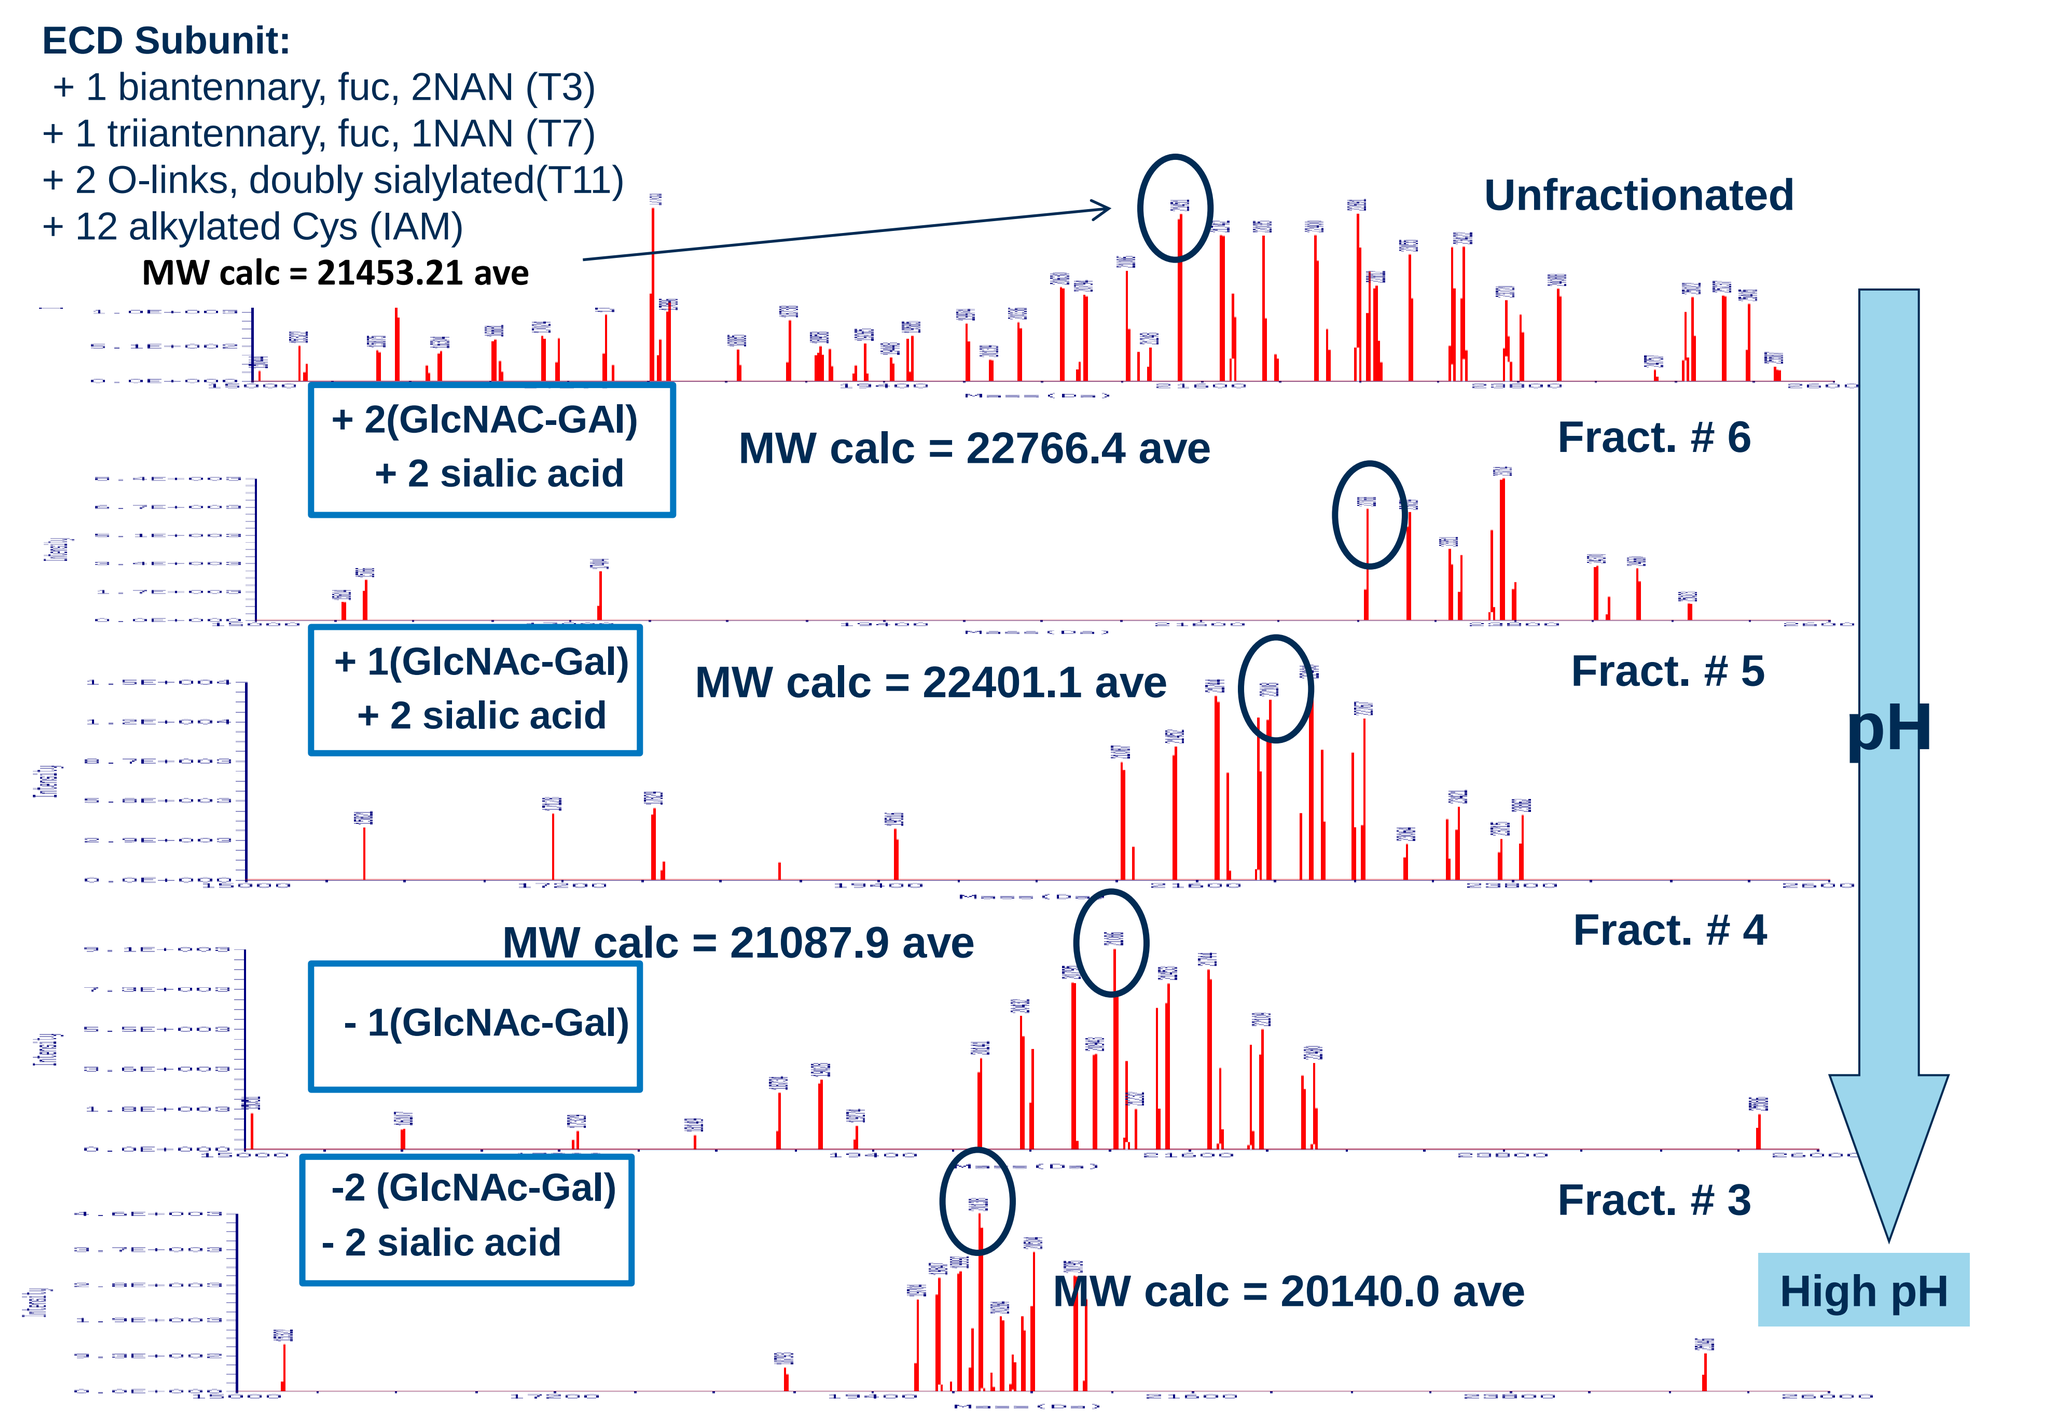

ECD Subunit:
+ 1 biantennary, fuc, 2NAN (T3)
+ 1 triiantennary, fuc, 1NAN (T7)
+ 2 O-links, doubly sialylated(T11)
+ 12 alkylated Cys (IAM)
MW calc = 21453.21 ave
Unfractionated
+ 2(GlcNAC-GAl)
+ 2 sialic acid
MW calc = 22766.4 ave
Fract. # 6
+ 1(GlcNAc-Gal)
+ 2 sialic acid
MW calc = 22401.1 ave
Fract. # 5
MW calc = 21087.9 ave
Fract. # 4
- 1(GlcNAc-Gal)
-2 (GlcNAc-Gal)
- 2 sialic acid
Fract. # 3
MW calc = 20140.0 ave
pH
High pH

Figure 10. Middle down LC/MS analysis of the ECD subunit from SAX fractions

The ECD subunit masses from the SAX fractions also shows that at the lower pH the glycan structures are more complex and have a grater amount of sialic acid.

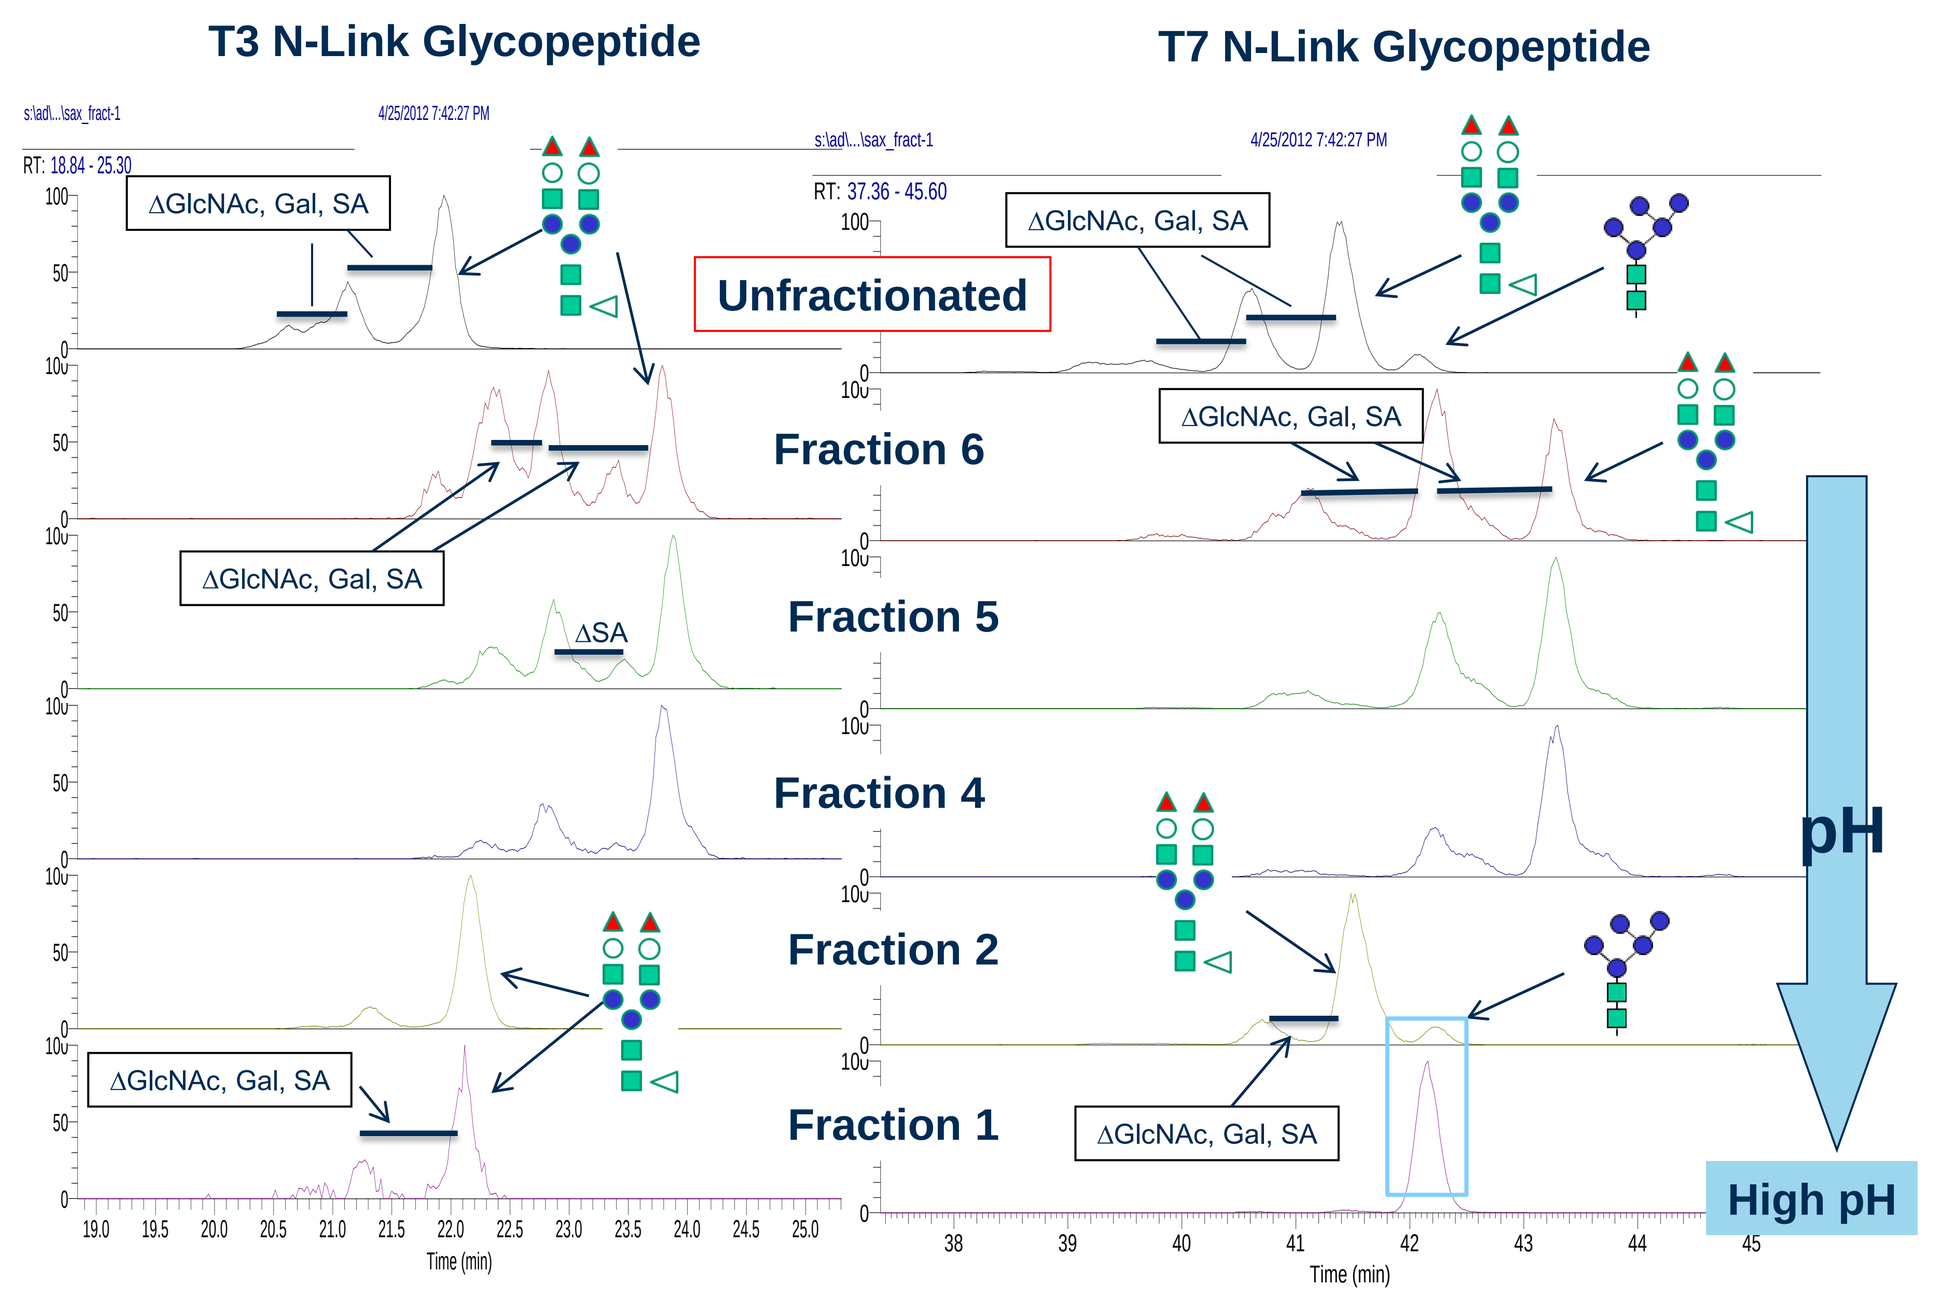

T3 N-Link Glycopeptide
T7 N-Link Glycopeptide
ΔGlcNAc, Gal, SA
ΔSA
Unfractionated
Fraction 6
Fraction 5
Fraction 4
Fraction 2
Fraction 1
pH
High pH
Time (min)

Figure 9. Peptide map analysis of the N-link glycosylations from SAX fractions

The lower pH fractions at both sites have glycosylations that have higher order branching and a greater amount of sialic acid. The majority of the higher pH fractions have glycosylations that are occupied with less complex biantennary and high mannose structures.

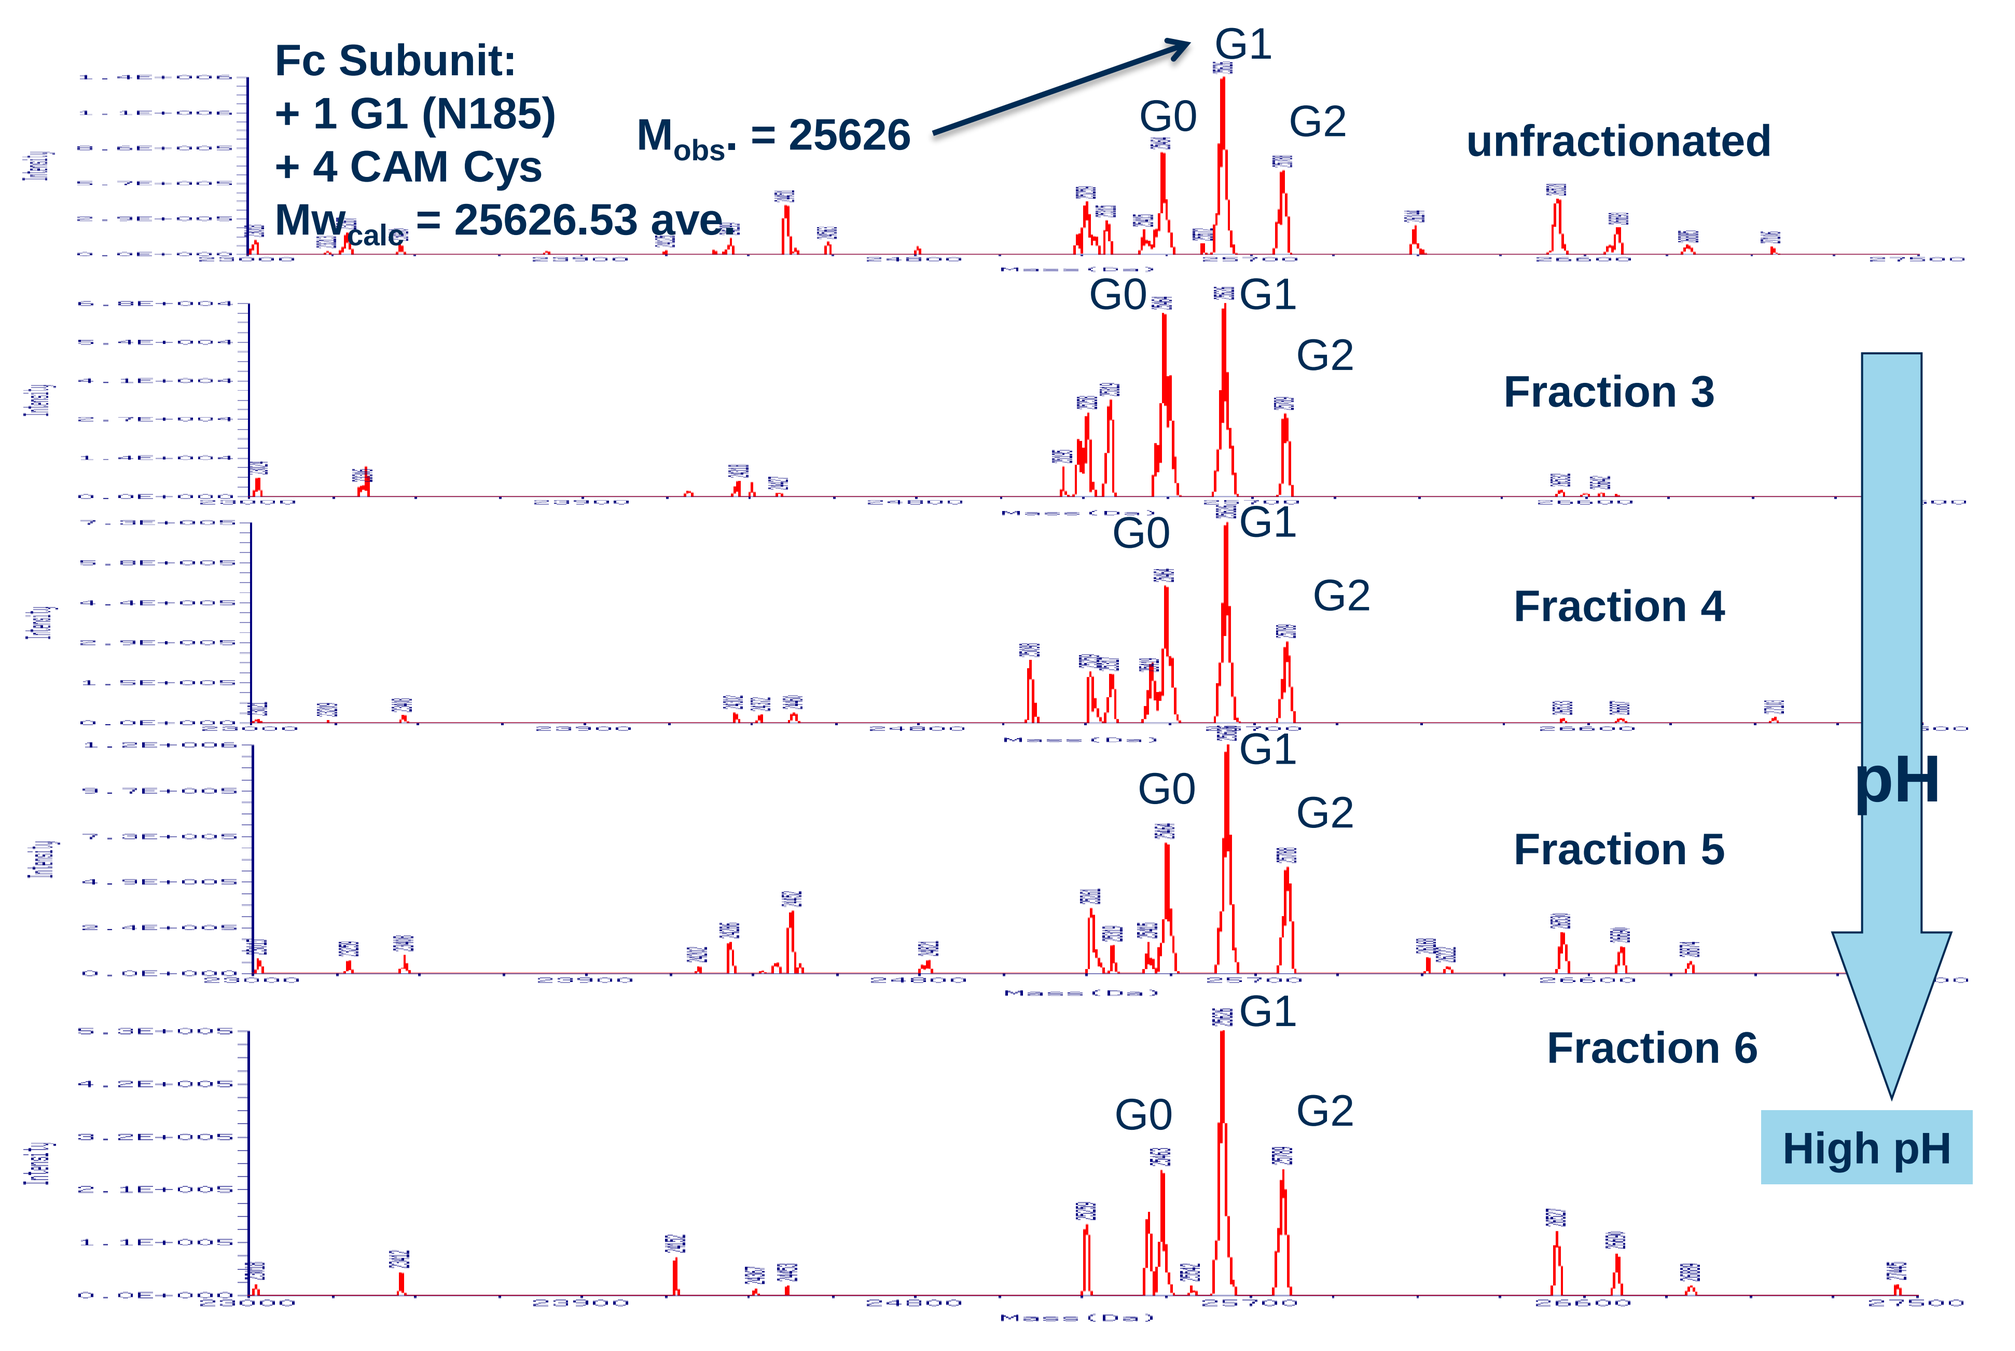

Fc Subunit:
+ 1 G1 (N185)
+ 4 CAM Cys
Mw_{calc} = 25626.53 ave.
M_{obs}. = 25626
G0
G1
G2
unfractionated
Fraction 3
Fraction 4
Fraction 5
Fraction 6
pH
High pH

Figure 11. Middle down LC/MS analysis of the Fc domain from the SAX fractions

This data is also showing that SAX fractionation has little effect on the Fc glycan distribution and the overall structure of the Fc domain.

Conclusions

1. Both of these fractionation techniques can be successfully developed into workflow platforms to structurally identify charge variants of highly complex glycoproteins
2. Both workflows show that at the lower pH there is an increasing complexity of glycans with higher order branching and more sialic acids
3. The subunit analysis of the Fc shows that there is little effect in the ratios of the Fc glycans in the separate fractions.
4. The N-link glycosylations of the ECD domain and the O-link glycosylations in the hinge region are the main contributors to the charge heterogeneity